UCL

Institute of Hepatology, UCL

Treatment of hyperammonaemia with OCR-002 (L-ornithine phenylacetate) reduces portal pressure by modulating hepatic NFkB and hepatic eNOS activity in cirrhotic rats

Rajeshwar P Mookerjee, Balasubramaniyan Vairappan, Vikram Sharma, Naina Shah, Abeba Habtesion, Nathan Davies, Rajiv Jalan

Liver Failure Group, Institute of Hepatology, University College London, UK.

Background

❖In astrocyte cultures, hyperammonaemia induces oxidative stress and NFkB and promotes inflammation (Norenberg 2008)

❖Hyperammonaemic animals manifest increased TNFα levels (Jalan 2007), and reduced bioavailability of brain nitric oxide (Felipo 2007), which can be corrected by reducing inflammation with ibuprofen.

❖In cirrhosis, severity of portal hypertension is associated with increased hepatic inflammation, reduced eNOS activity and increased endogenous NOS inhibitors, Caveolin-1 and asymmetric-dimethylarginine (ADMA) (Mookerjee 2007)

❖OCR-002 is a novel salt made up of equimolar concentration of L-ornithine and Phenylacetate, shown to be effective in:

- reducing ammonia and brain water in cirrhotic rats (Davies et al. 2009)
- attenuating rise in ammonia and intracranial pressure in devascualarised acute liver failure pigs (Ytrebo et al, 2009)

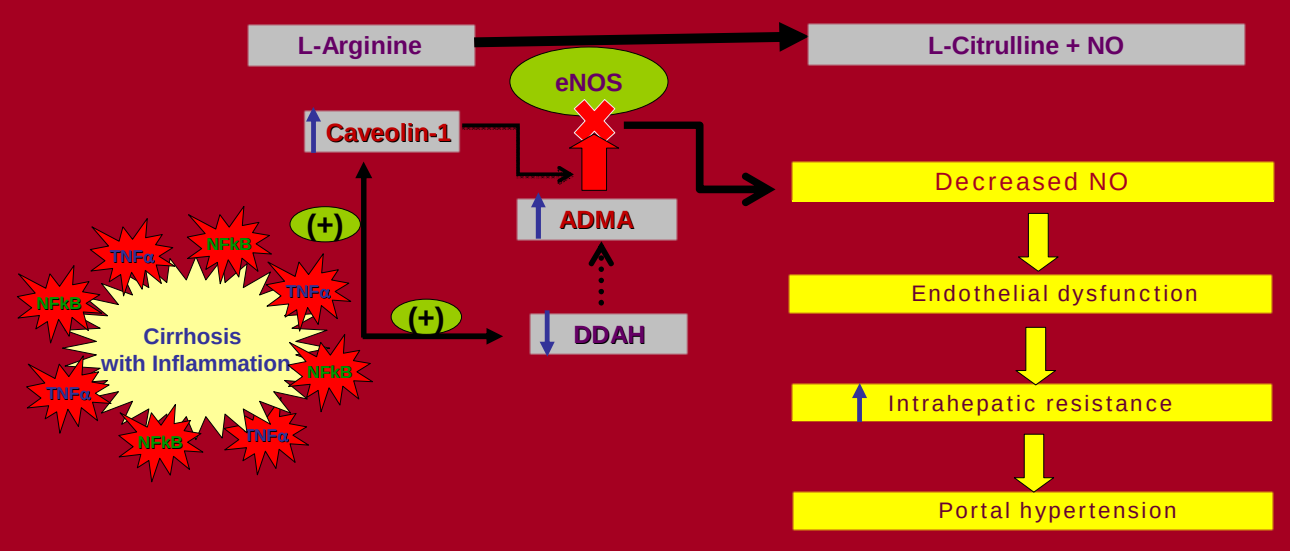
L-Arginine
L-Citrulline + NO
eNOS
Caveolin-1
ADMA
DDAH
(+)
(+)
Cirrhosis
with Inflammation
TNFα
NFkB
Decreased NO
Endothelial dysfunction
Intrahepatic resistance
Portal hypertension

Aims

❖This study was designed to test the hypothesis that reducing ammonia concentrations with OCR-002 (L-ornithine phenylacetate) will:

- reduce systemic and hepatic inflammation
- increase intrahepatic NO availability through effects on the NO regulatory pathways of caveolin-1 and DDAH-ADMA
- thereby reduce portal pressure

Methods

<u>Animals:</u> Four weeks after BDL and sham surgery in Sprague-Dawley rats, BDL rats were given i.p. OCR-002 3g/kg twice a day or vehicle alone (n=14/group), and treated for 5 days .

<u>Measurements:</u> After the 5th treatment day, all rats underwent direct portal pressure measurement under anaesthesia prior to sacrifice. Plasma and liver tissue was harvested for subsequent analysis:

- ➢ Plasma ammonia and biochemistry were measured using a Cobas-Integra analyser
- ➢ Plasma TNFα was measured by FACS cytometric bead array
- ➢ eNOS activity was determined radiometrically
- ➢ Protein expression for eNOS, DDAH-1, caveolin-1, and NFkB were measured using standard Western Blotting techniques.

Results

Parameters	Sham	BDL	BDL+OP
Ammonia (µmol/L)	55.86±4.05	222.3±24.01**	127.1±47.8†
ALT (U/L)	26.88±2.61	159.8±10.81**	108.3±5.66†
Bilirubin (µmol/L)	32.52±2.43	304.9±25.97**	267.7±14.5NS
Creatinine (µmol/L)	23.07±0.94	40.88±2.37*	32.38±1.44§
TNF-α (pg/ml)	121.9±46.4	822.9±203.1*	270.5±59.4§

Table - Effect of OCR-002 (OP) on arterial ammonia and plasma biochemistry of BDL rats. Statistical significance was calculated using student t' test Mann Witney comparisons test. * - P<0.01 versus sham; ** - P<0.001 versus sham; † - P<0.01 versus BDL; § - P<0.05 versus BDL; NS-no significance.

a) Hepatic eNOS activity

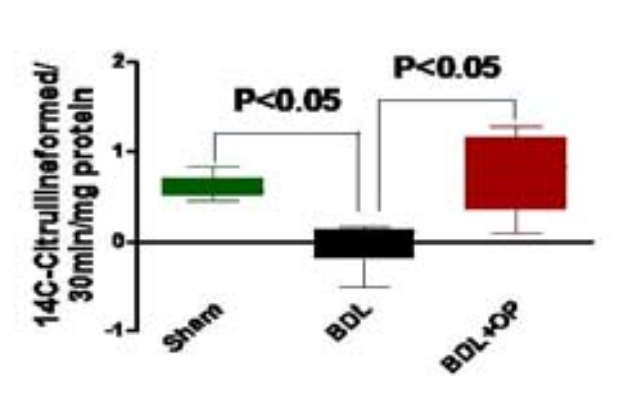
P<0.05
P<0.05
14C-Citrullineformed/
30min/mg protein
2
1
0
-1
Sham
BDL
BDL+OP

b) Hepatic eNOS protein expression

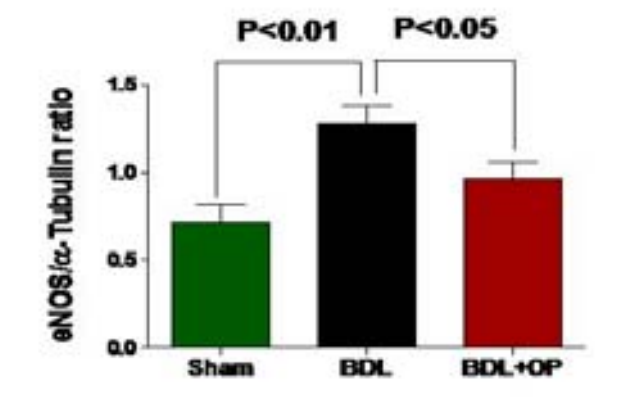
P<0.01
P<0.05
eNOS/α-Tubulin ratio
1.5
1.0
0.5
0.0
Sham
BDL
BDL+OP

eNOS activity is significantly decreased in BDL animals compared to sham (p<0.05) despite increased eNOS protein expression (p<0.01). Following treatment with OCR-002, eNOS activity reverts to sham levels, with similar normalisation of eNOS protein expression (p<0.05)

c) Hepatic DDAH-1 protein expression

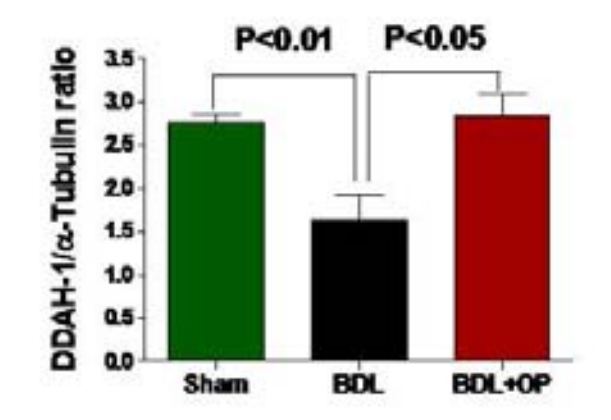
P<0.01
P<0.05
DDAH-1/α-Tubulin ratio
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
Sham
BDL
BDL+OP

DDAH-1 protein expression is significantly higher in sham animals compared to BDL (p<0.01). Following OCR-002 administration to BDL rats, DDAH-1 expression increases significantly and reverts to sham levels (p<0.05)

d) Hepatic NFkB protein expression

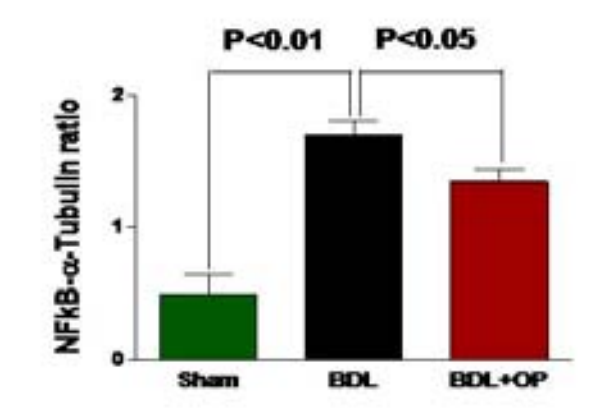
P<0.01
P<0.05
NFkB-α-Tubulin ratio
2
1
0
Sham
BDL
BDL+OP

Protein expression of NFkB is significantly elevated in BDL rats compared to sham (p<0.01). OCR-002 treatment produces a marked reduction in NFkB expression compared to BDL (p=0.05).

e) Hepatic caveolin-1 protein expression

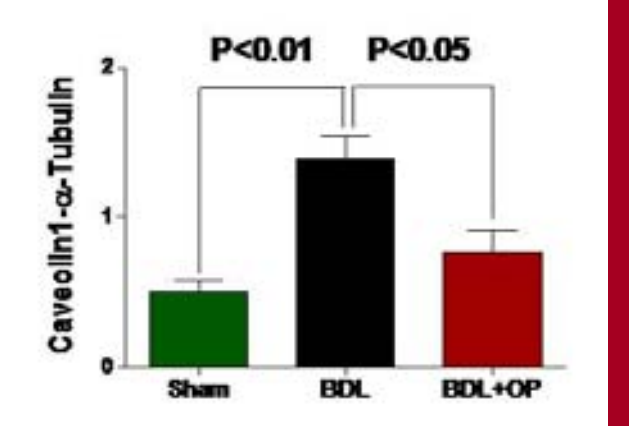
P<0.01
P<0.05
Caveolin1-α-Tubulin
2
1
0
Sham
BDL
BDL+OP

The expression of caveolin-1 is significantly elevated in BDL rats compared to sham (p<0.01). Following intervention with OCR-002 Caveolin-1 expression decreases significantly (p<0.05) compared to BDL alone.

f) Portal pressure

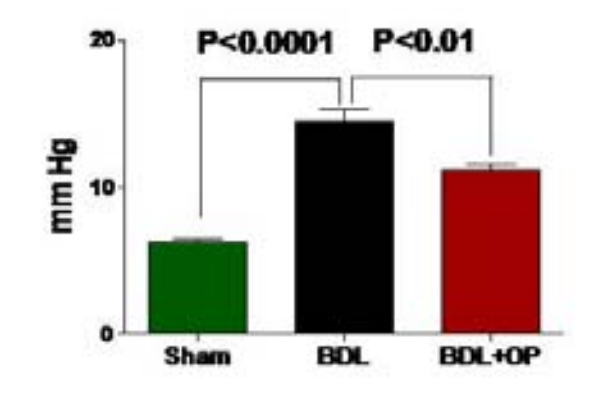
P<0.0001
P<0.01
mm Hg
20
10
0
Sham
BDL
BDL+OP

Portal pressure is significantly increased in BDL rats compared to sham (p<0.0001). OCR-002 treatment results in a 30% reduction of portal pressure as compared to BDL (P<0.01)

Conclusions

➢ This study demonstrates for the first time that treatment of hyperammonaemia with OCR-002 in a clinically relevant model of cirrhosis reduces portal pressure significantly by:

- Reducing systemic, hepatic and NFkB inflammatory signalling
- Restoring hepatic eNOS activity by favourably modulating the inhibitory pathways of DDAH-ADMA and caveolin-1

➢ Our data provides the rationale for evaluating OCR-002 in the treatment of portal hypertension.

Acknowledgement: Ornithine benzeneacetate, supplied by Ocera Therapeutics, San Diego, CA